Unlocking precision: a next-generation multi-specific CD3 Switch-DARPin with enhanced function to tackle current limitations of T cell engagers in ovarian cancer

Joanna Robinson, Matteo Bianchi, Mischa Müller, Guillaume Ems, Chloé Friang, Yvonne Grübler, Christel Herzog, Maria Paladino, Stefanie Riesenberg, Albulena Toska, Denis Villemagne, Justin Walter, Valérie Calabro, Marcel Walser, Anne Goubier and Marcela Guzman Ayala; Molecular Partners AG, Zürich-Schlieren, Switzerland

Poster # 842

2024 SITC Annual Meeting

Introduction

Ovarian cancer (OC) is characterized by a lack of durable response and the absence of specific, targeted therapy leads to resistance. Immunotherapy has had limited success, attributed to the absence of clean tumor-associated antigens (TAAs), presence of dysfunctional T cells, and an immunosuppressive tumor microenvironment. To address all these challenges in one molecule, we boosted our mesothelin (MSLN) x CD3 DARPin T cell engager with a CD2-engaging domain for co-stimulation and engineered a second "switchable" TAA-targeting DARPin (EpCAM) with a CD3-mask to ensure activation ONLY in the presence of these TAAs – further referred to as CD2/CD3 Switch.

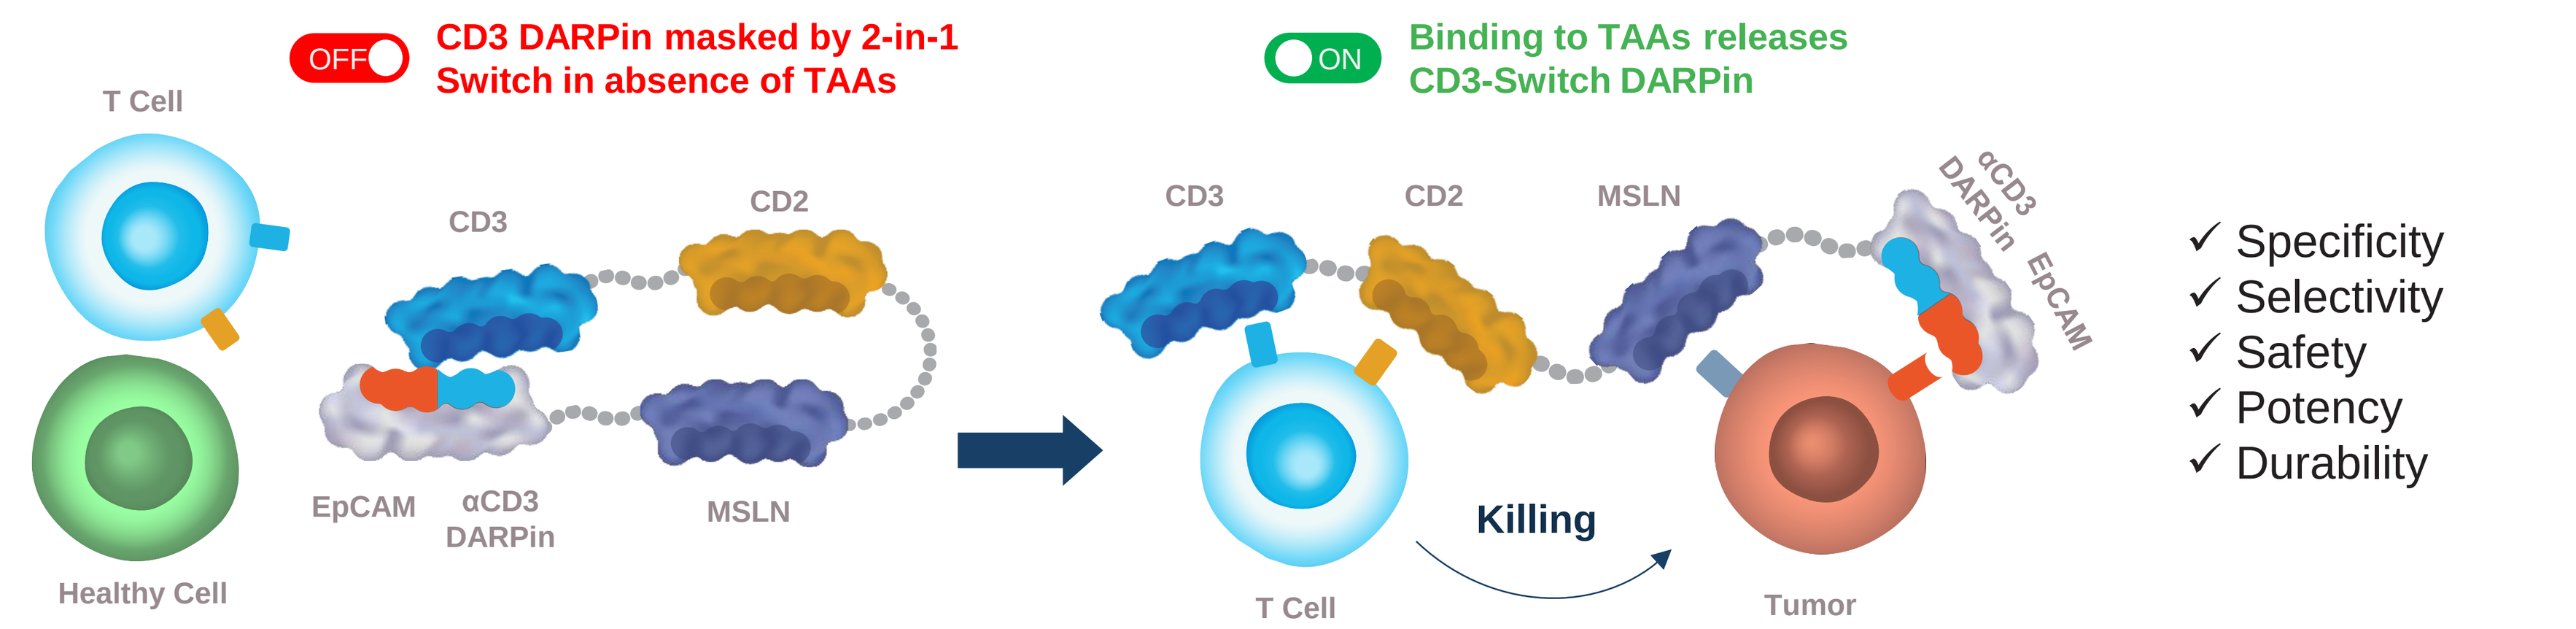
OFF CD3 DARPin masked by 2-in-1 Switch in absence of TAAs
ON Binding to TAAs releases CD3-Switch DARPin
T Cell
Healthy Cell
CD3
CD2
EpCAM
αCD3 DARPin
MSLN
Killing
Tumor
✓ Specificity
✓ Selectivity
✓ Safety
✓ Potency
✓ Durability

EpCAM & MSLN are highly co-expressed in OC tumors

RNAseq data from human tissue biopsies, cancer cells and T cells

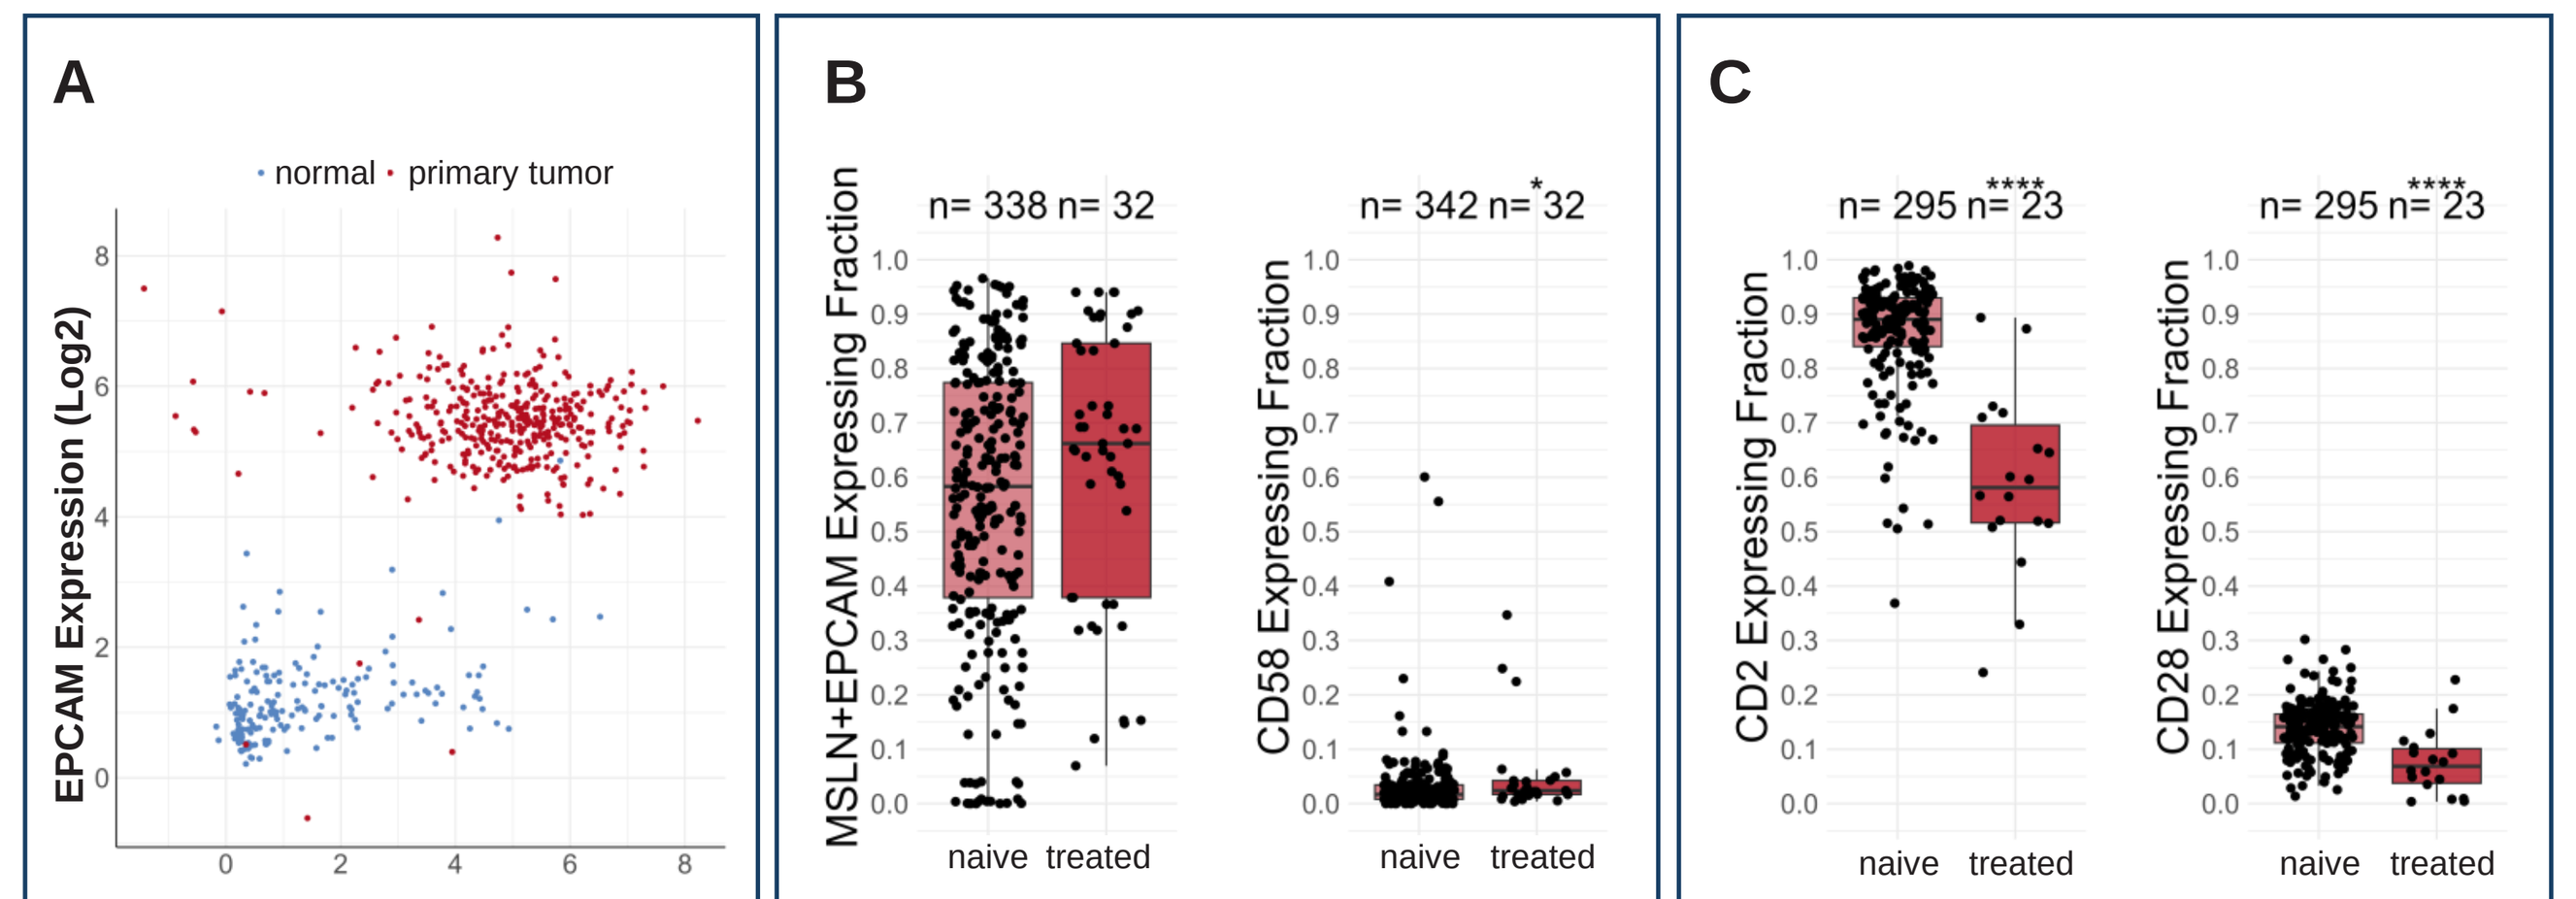
A
normal · primary tumor
EPCAM Expression (Log2)
MSLN Expression (Log2)
B
MSLN+EPCAM Expressing Fraction
CD58 Expressing Fraction
n= 338 n= 32
n= 342 n= 32
naive treated
C
CD2 Expressing Fraction
CD28 Expressing Fraction
n= 295 n= 23
naive treated

(A) Tissue sample biopsies from TCGA and GTEx were analyzed for MSLN and EpCAM expression (RNAseq, bulk).
(B,C) Public single-cell RNAseq data were curated and re-analyzed in-house. The fraction of cells expressing the gene of interest was determined.
(n= number of samples; unpaired Wilcoxon test).
Panel A: tissue biopsies; Panel B: cancer cells; Panel C: T cells.

In-house curated data show that ~60% of untreated primary tumours co-express MSLN and EpCAM, while this is rare in healthy tissue. CD58, the ligand for CD2, is often down-regulated in OC, whilst >60% of infiltrating T cells express CD2, compared to <15% for CD28.

CD3 Mask prevents T cell activation in the absence of TAAs

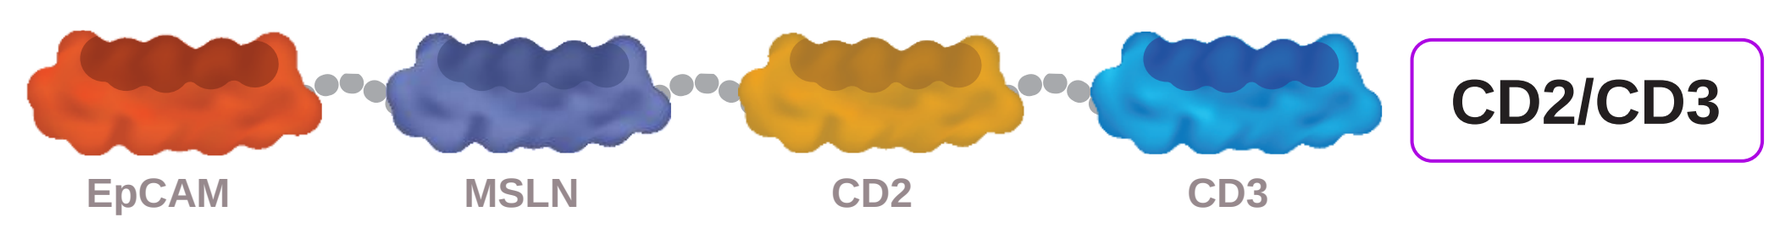
EpCAM
MSLN
CD2
CD3
CD2/CD3

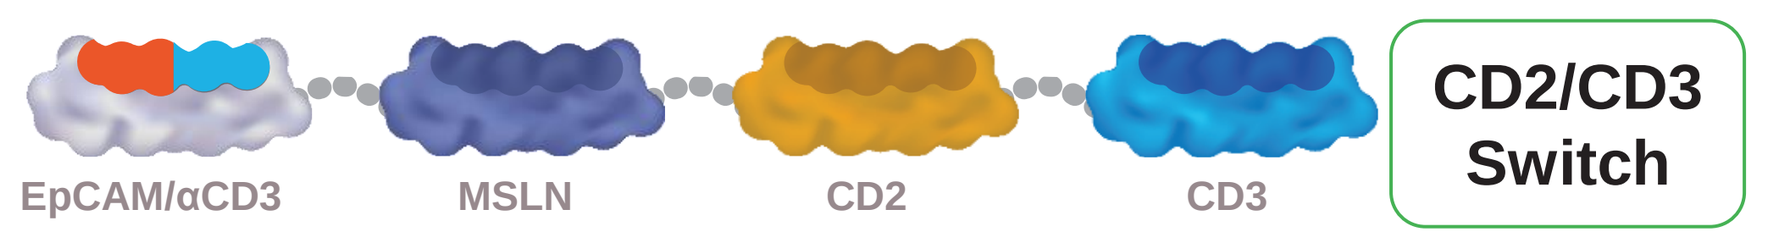
EpCAM/αCD3
MSLN
CD2
CD3
CD2/CD3 Switch

T cell Activation

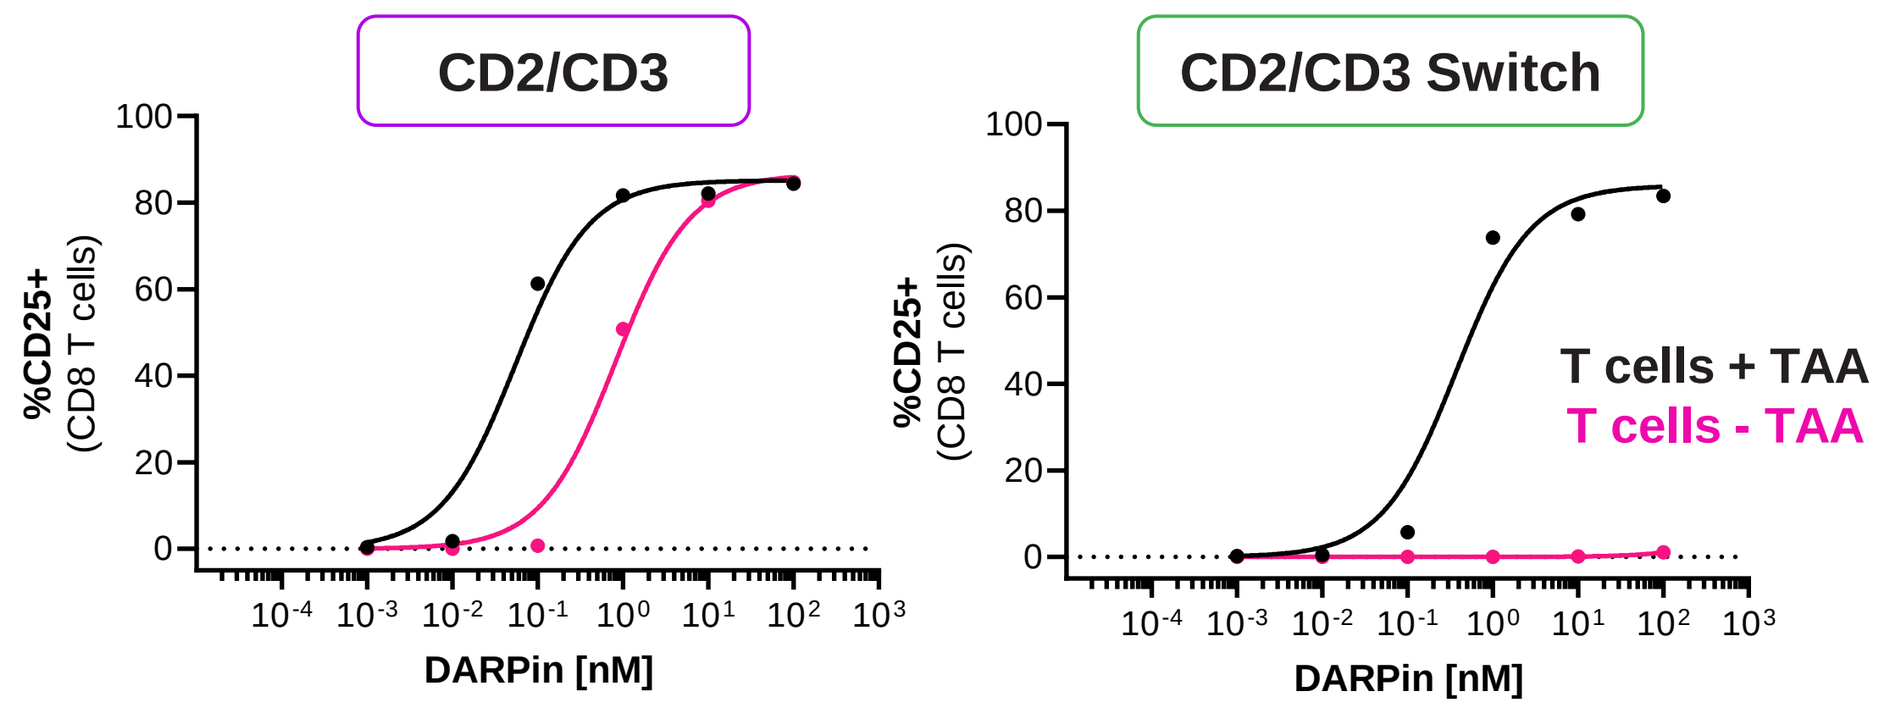
CD2/CD3
CD2/CD3 Switch
%CD25+ (CD8 T cells)
T cells + TAA
T cells - TAA
DARPin [nM]

Pan T cells co-cultured with or without target cells (OVCAR-3 WT, 5:1). Activation (CD25) of CD8 T cells measured by flow cytometry after 48h.

T cell Cytotoxicity

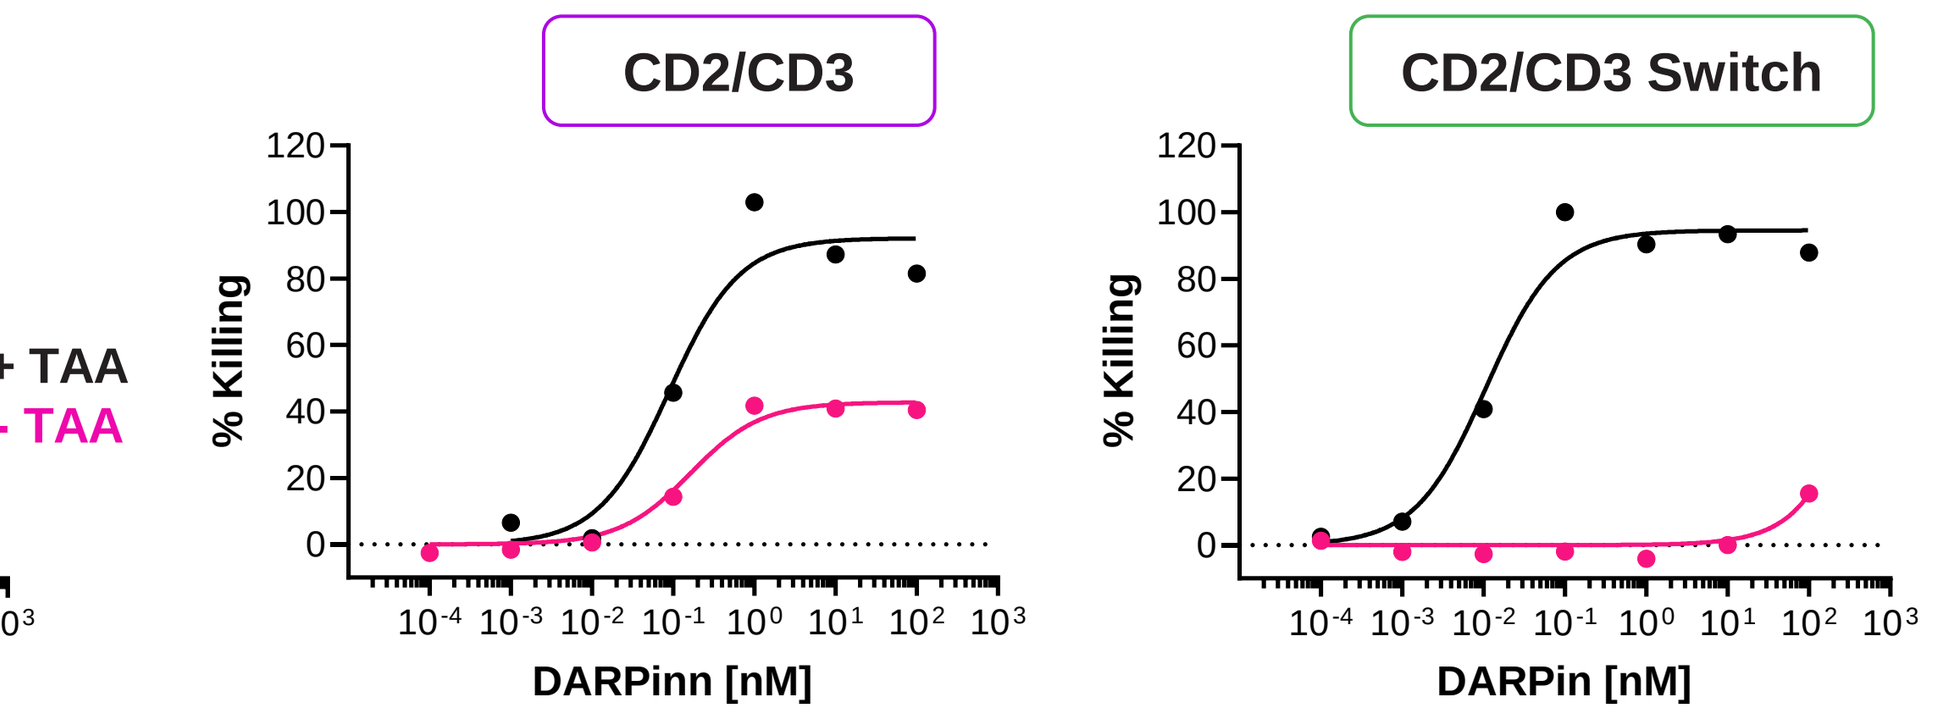
CD2/CD3
CD2/CD3 Switch
% Killing
DARPinn [nM]
DARPin [nM]

Pan T cells co-cultured with CHO WT or CHO-EpCAMxMSLN (1:1). LDH is measured in the supernatant after 48h and normalized to 100% killing.

CD2/CD3 Switch induces tumor regression in humanized mice

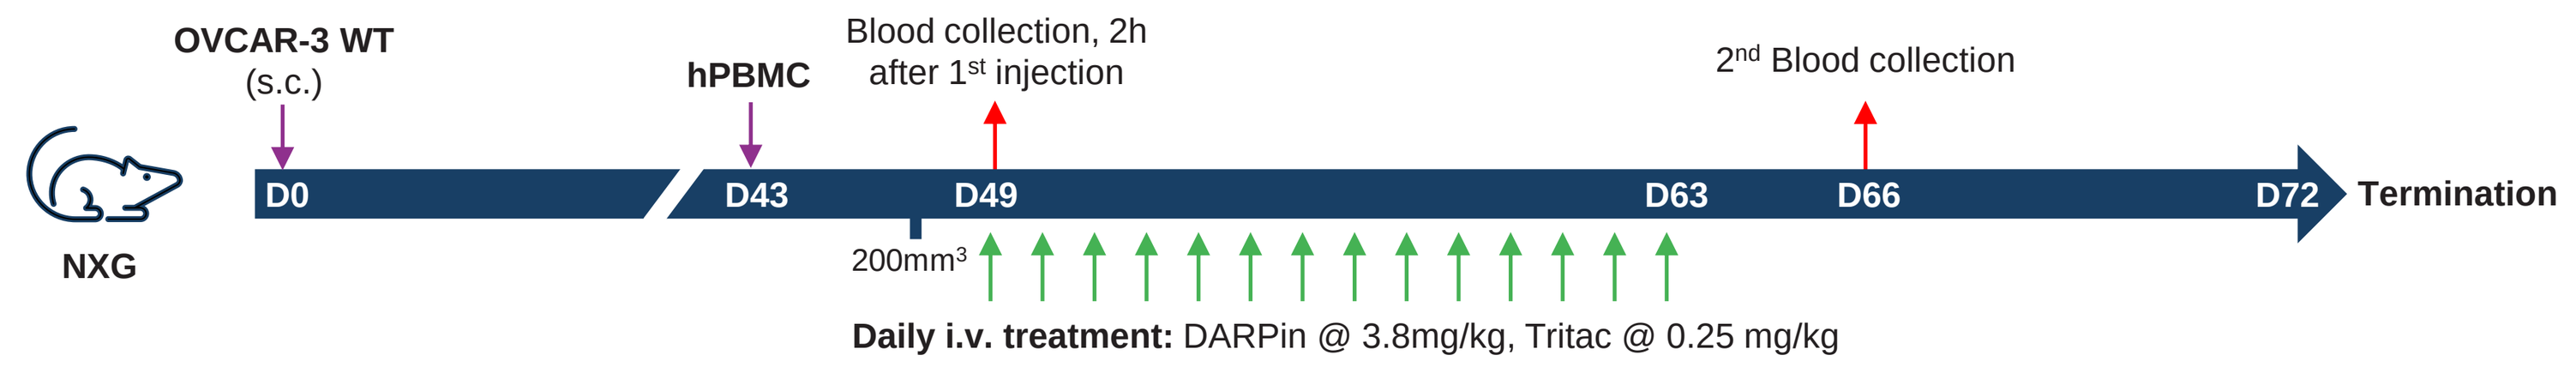
OVCAR-3 WT (s.c.)
hPBMC
Blood collection, 2h after 1st injection
2nd Blood collection
NXG
D0
D43
D49
D63
D66
D72
Termination
200mm³
Daily i.v. treatment: DARPin @ 3.8mg/kg, Tritac @ 0.25 mg/kg

Tumor growth curves of OVCAR-3 s.c. model

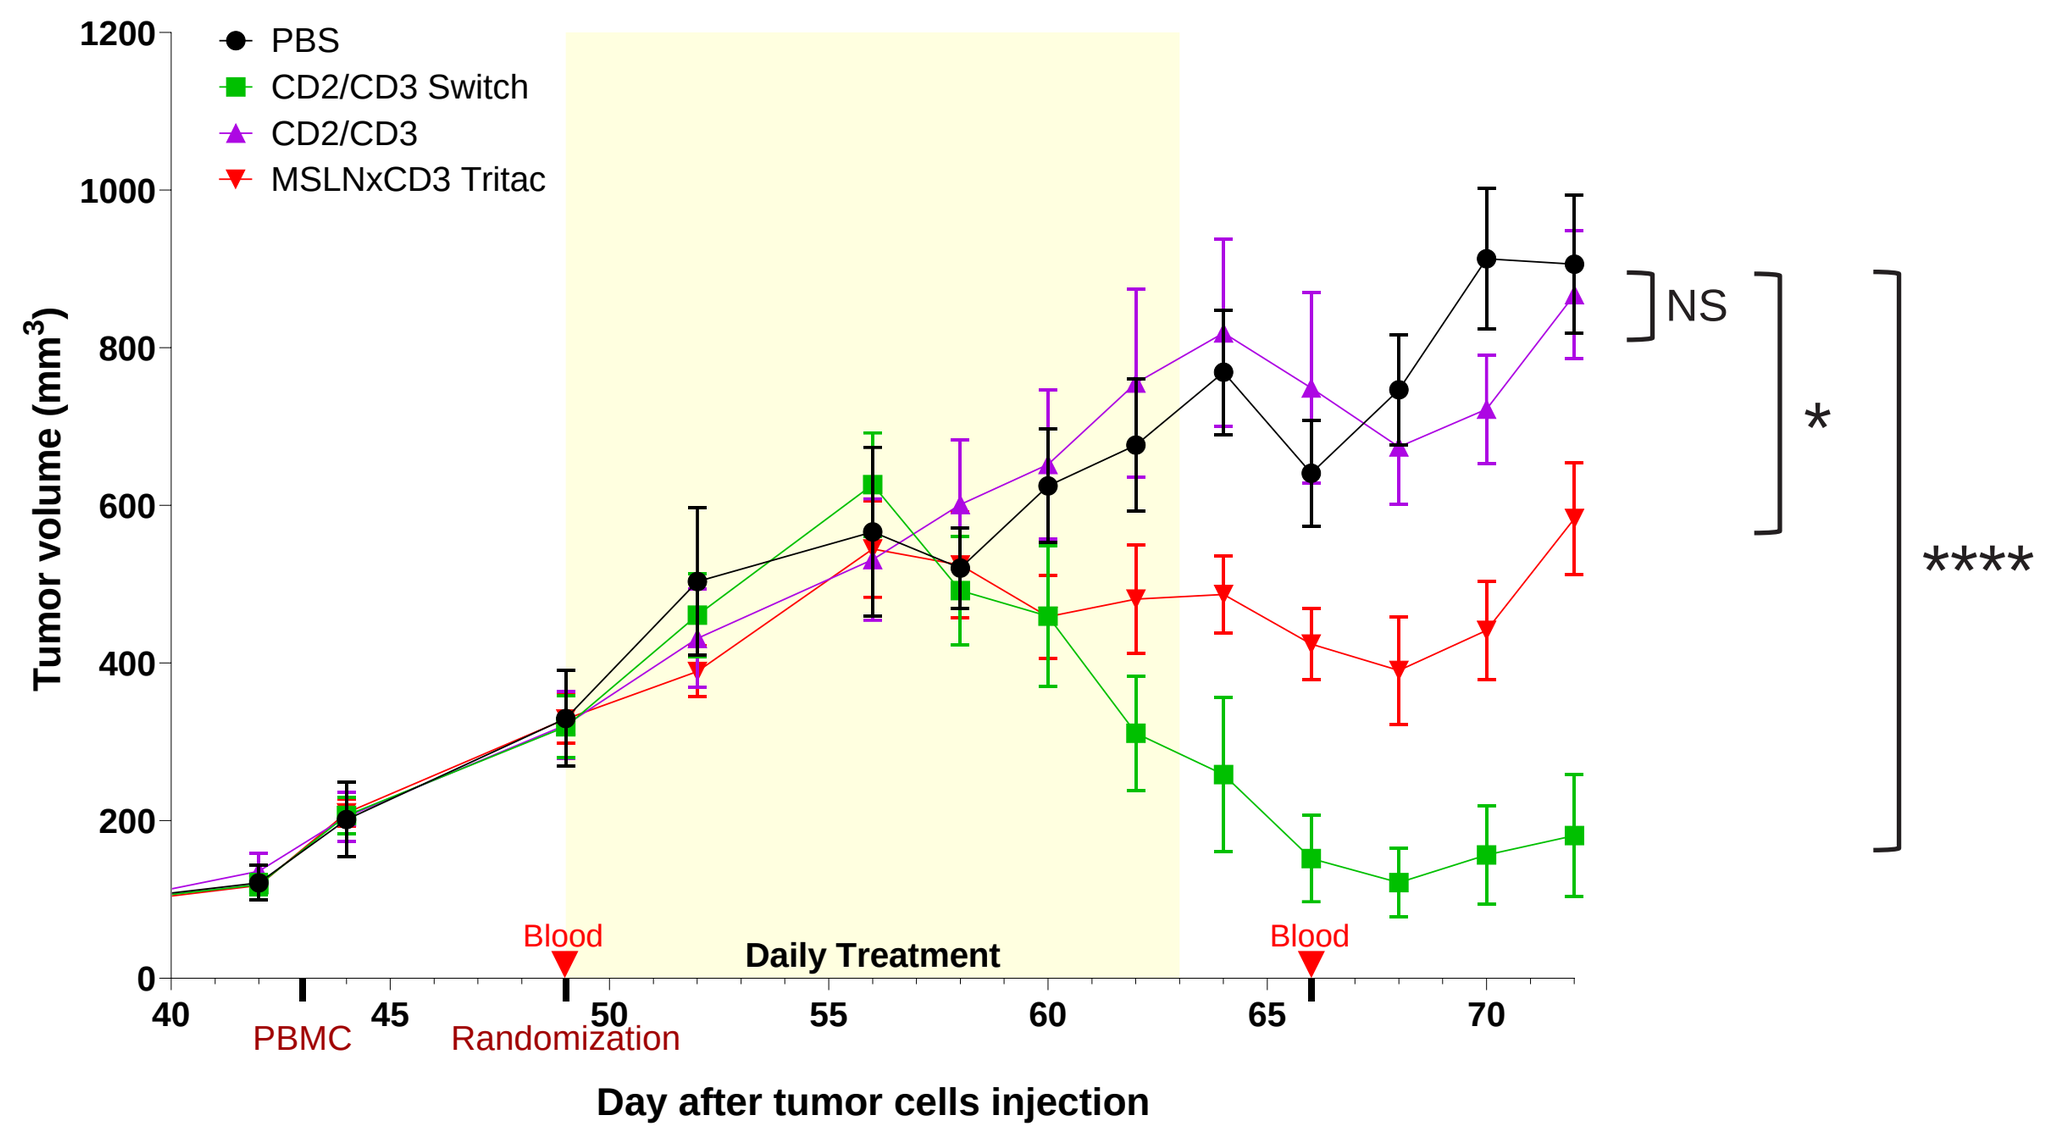
PBS
CD2/CD3 Switch
CD2/CD3
MSLNxCD3 Tritac
Tumor volume (mm³)
Daily Treatment
Blood
PBMC
Randomization
Day after tumor cells injection
NS
*

10 mice/group, 2 PBMC donors engrafted at 100mm³, average +/- SEM, two-way ANOVA

Human CD45+ in blood on Day 66

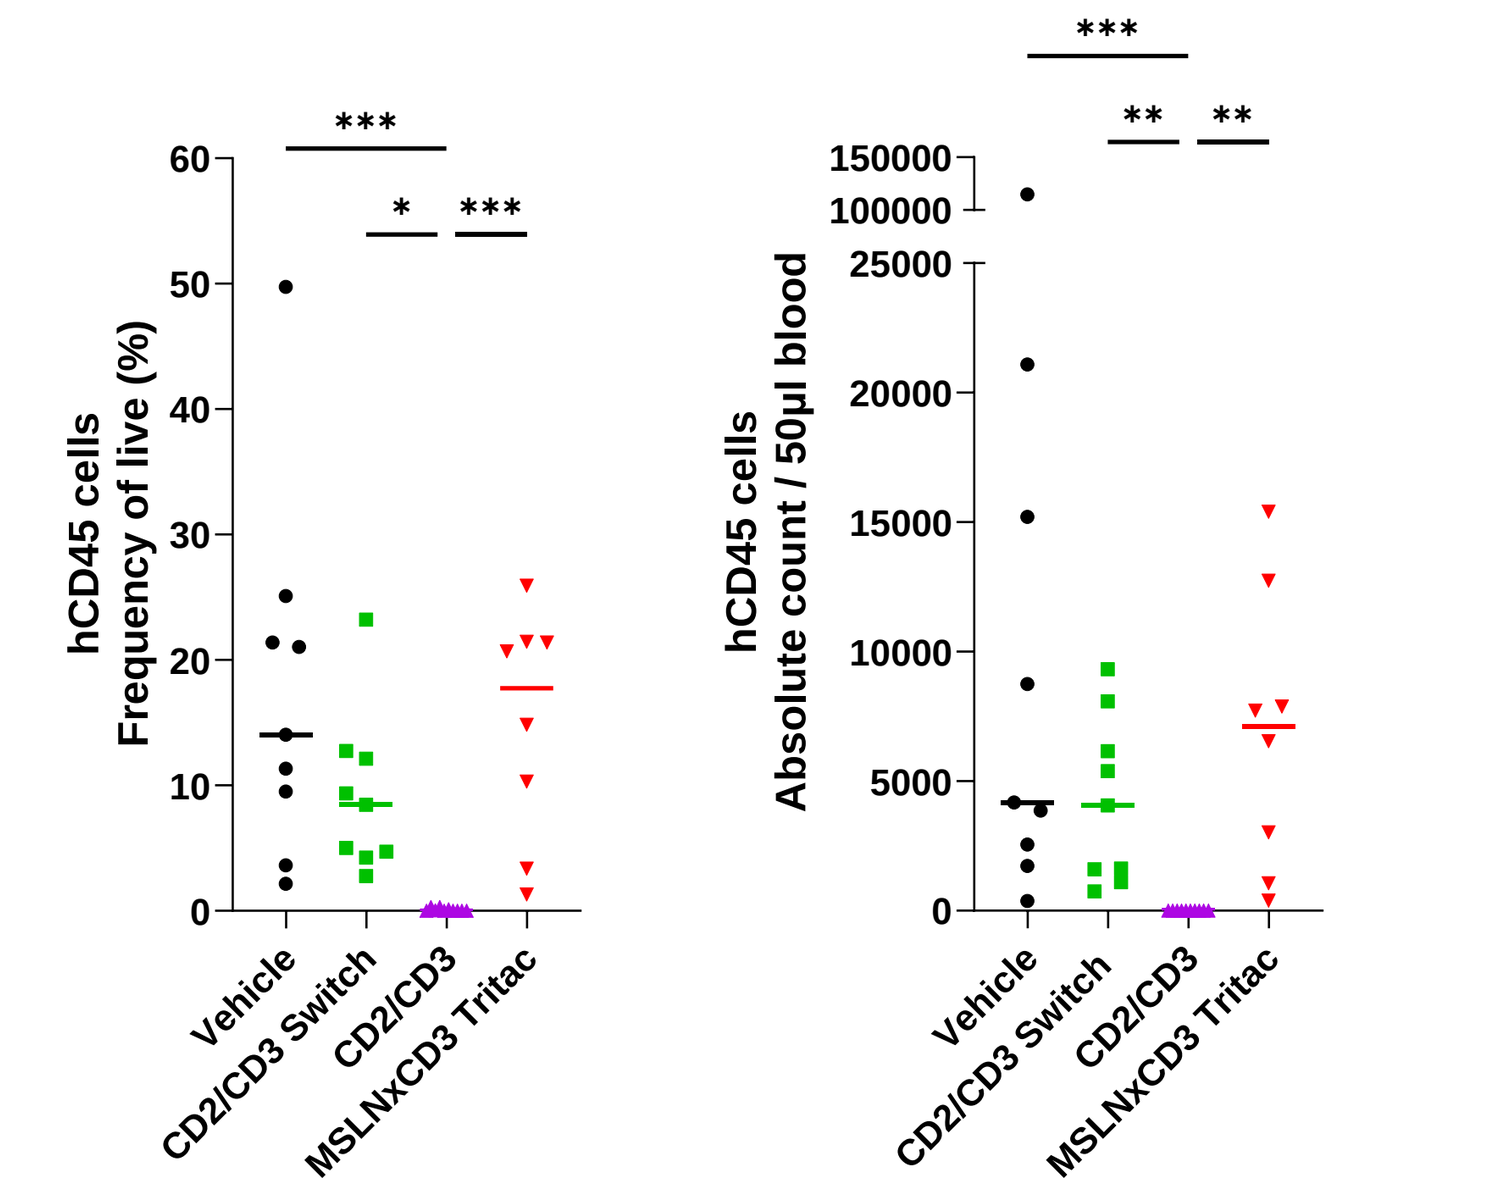
hCD45 cells Frequency of live (%)
hCD45 cells Absolute count / 50µl blood
Vehicle
CD2/CD3 Switch
CD2/CD3
MSLNxCD3 Tritac

Kruskal-Wallis test for all mice (median)*

CD2/CD3 Switch induces significant tumor regression. Masking of CD3 ensures activation specifically in the tumor, whereas unmasked CD2/CD3 is not efficacious, likely due to loss of CD45+ T cells as a result of non-specific T cell activation in the periphery.

Masking CD3 may reduce risk of CRS providing a better safety profile

Cytokines measured in mice blood 2h after first dose

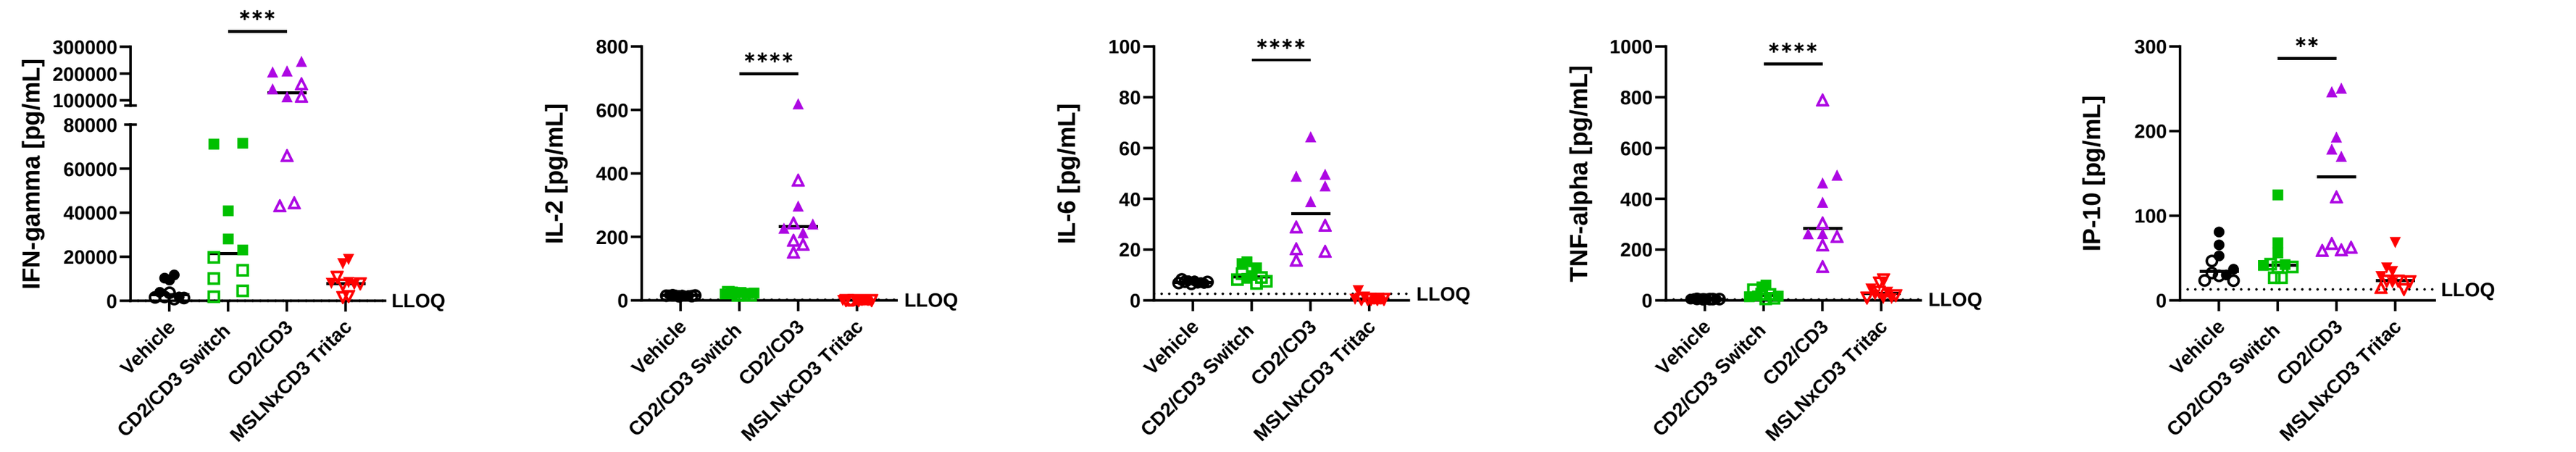
IFN-gamma [pg/mL]
IL-2 [pg/mL]
IL-6 [pg/mL]
TNF-alpha [pg/mL]
IP-10 [pg/mL]
LLOQ
Vehicle
CD2/CD3 Switch
CD2/CD3
MSLNxCD3 Tritac

*Serum from blood taken at 2h post first injection was analyzed using MSD U-plex system. Significance shown with t-test **

CD2/CD3 Switch shows significantly lower cytokine levels shortly after first administration, indicating that masking CD3 could reduce the risk of cytokine release syndrome (CRS).

Cytokine release from a human whole blood assay

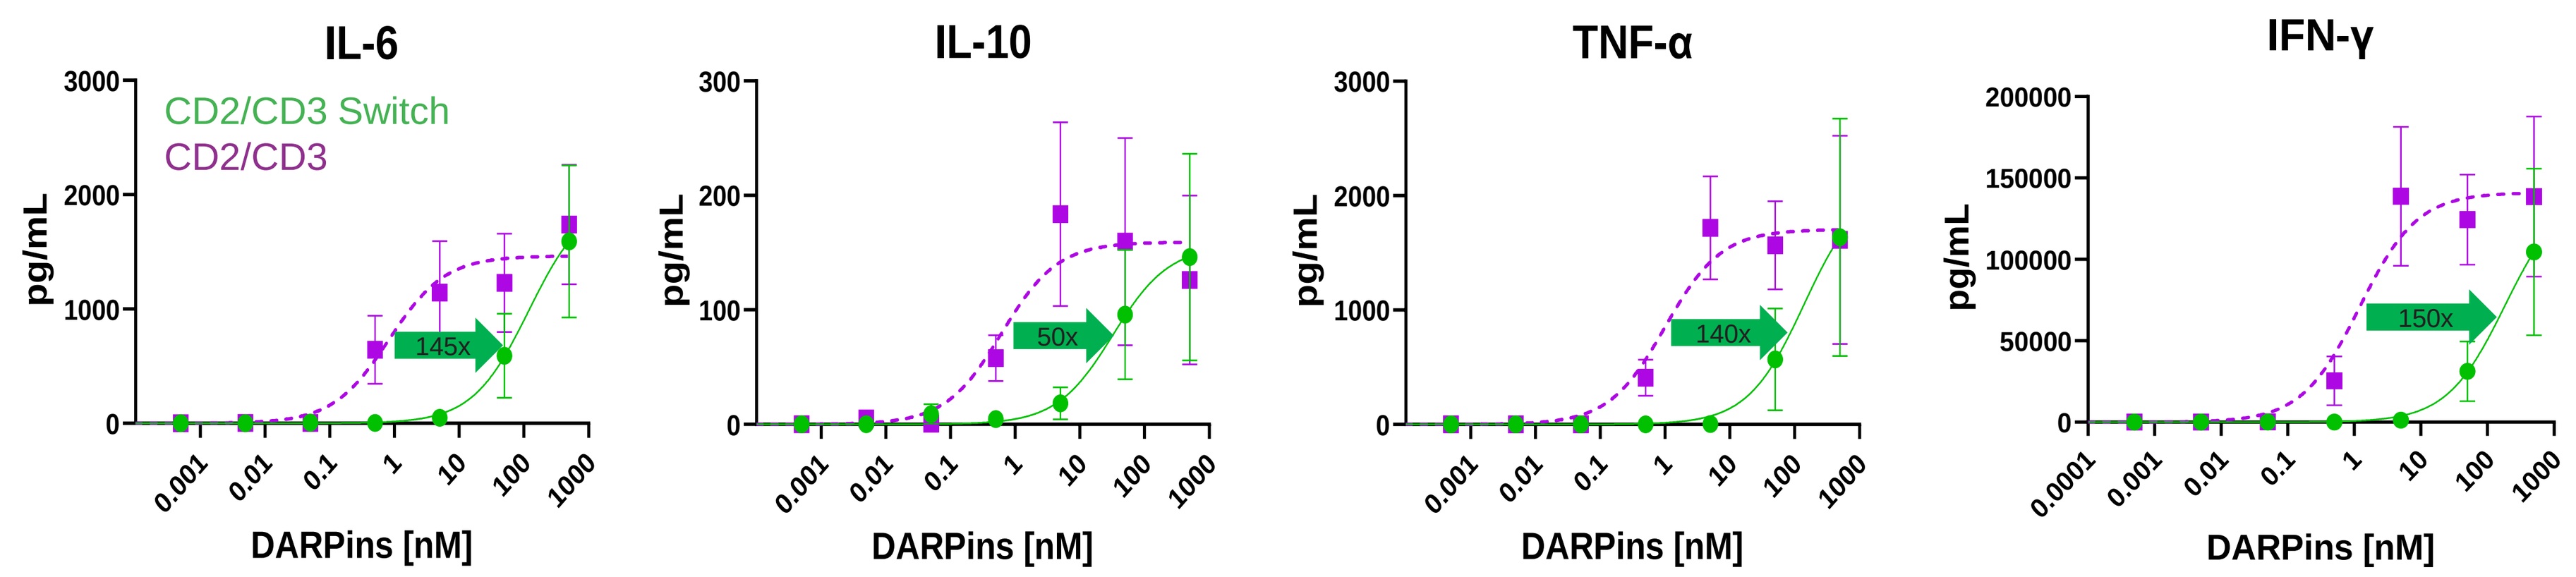
IL-6
IL-10
TNF-α
IFN-γ
CD2/CD3 Switch
CD2/CD3
pg/mL
145x
50x
140x
150x
DARPins [nM]

Whole blood from 3 donors incubated 24h with DARPin. Serum taken for cytokine analysis by MSD U-plex. Average +/- SEM.

CD2/CD3 DARPin induces non-TAA-specific cytokine release in human whole blood. CD2/CD3 Switch DARPin reduces non-specific cytokine release.

CD2 co-stimulation prevents loss of activity against CD58 KO cells

Reporter cell activation

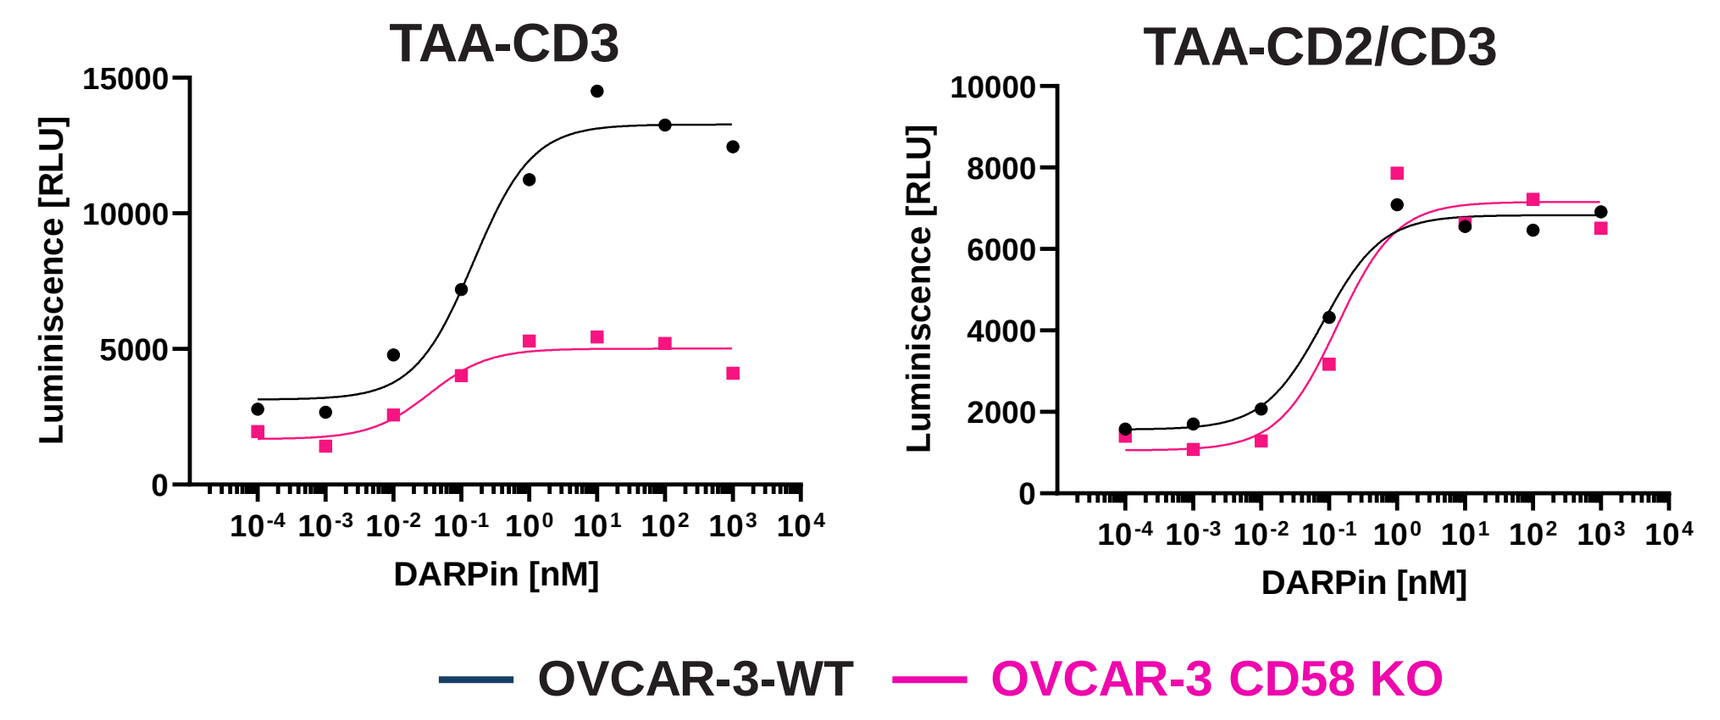
TAA-CD3
TAA-CD2/CD3
Luminiscence [RLU]
DARPin [nM]
OVCAR-3-WT
OVCAR-3 CD58 KO

Absence of CD58 on target cells results in reduced efficacy of the CD3 DARPin whereas CD2/CD3 co-stimulation maintains efficacy, thus compensating for the lack of CD58.

Jurkat IL-2 cells cultured 1:1 with OVCAR-3 WT or CD58 KO cells over 24h. Activation measured by luminescence.

CD2 co-stimulation enables continued polyfunctionality of T cells

T cells repetitively stimulated with CD2/CD3 DARPins maintain cytotoxic capacity whereas T cells cultured in the presence of DARPins lacking CD2 show reduced capacity of target cell killing. T cells repetitively exposed to CD2/CD3 DARPin maintain a more activated phenotype than to CD3 alone.

Repetitive stimulation scheme

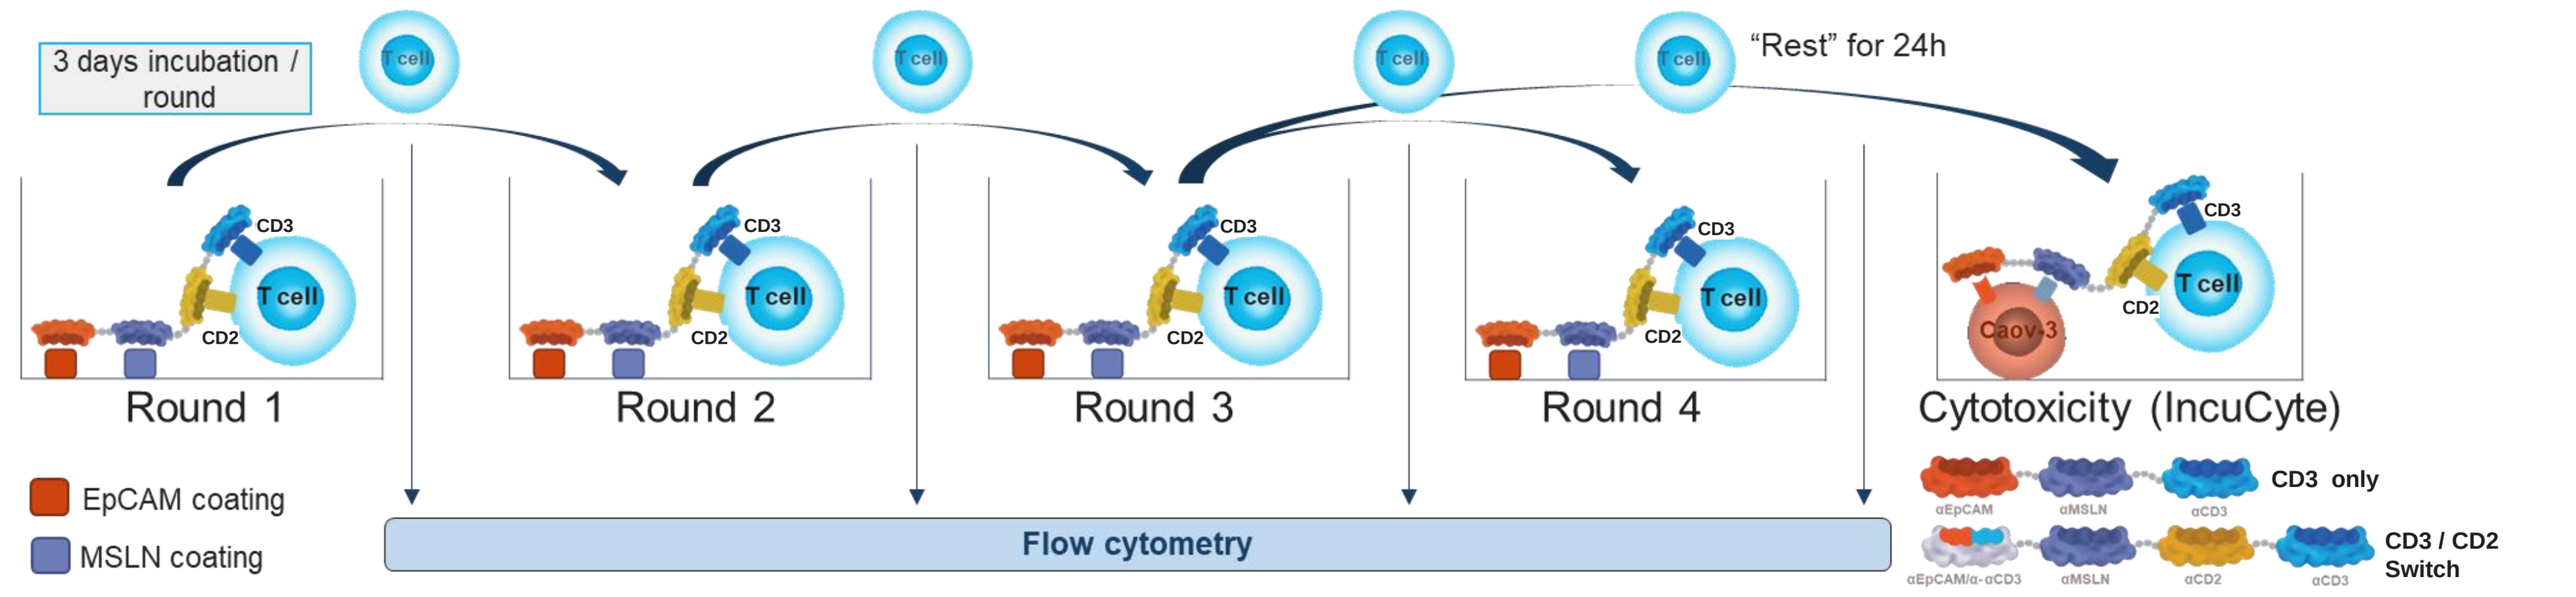
3 days incubation / round
"Rest" for 24h
Round 1
Round 2
Round 3
Round 4
Cytotoxicity (IncuCyte)
EpCAM coating
MSLN coating
Flow cytometry
CD3 only
CD3 / CD2 Switch

Phenotype of T cells after repetitive stimulation

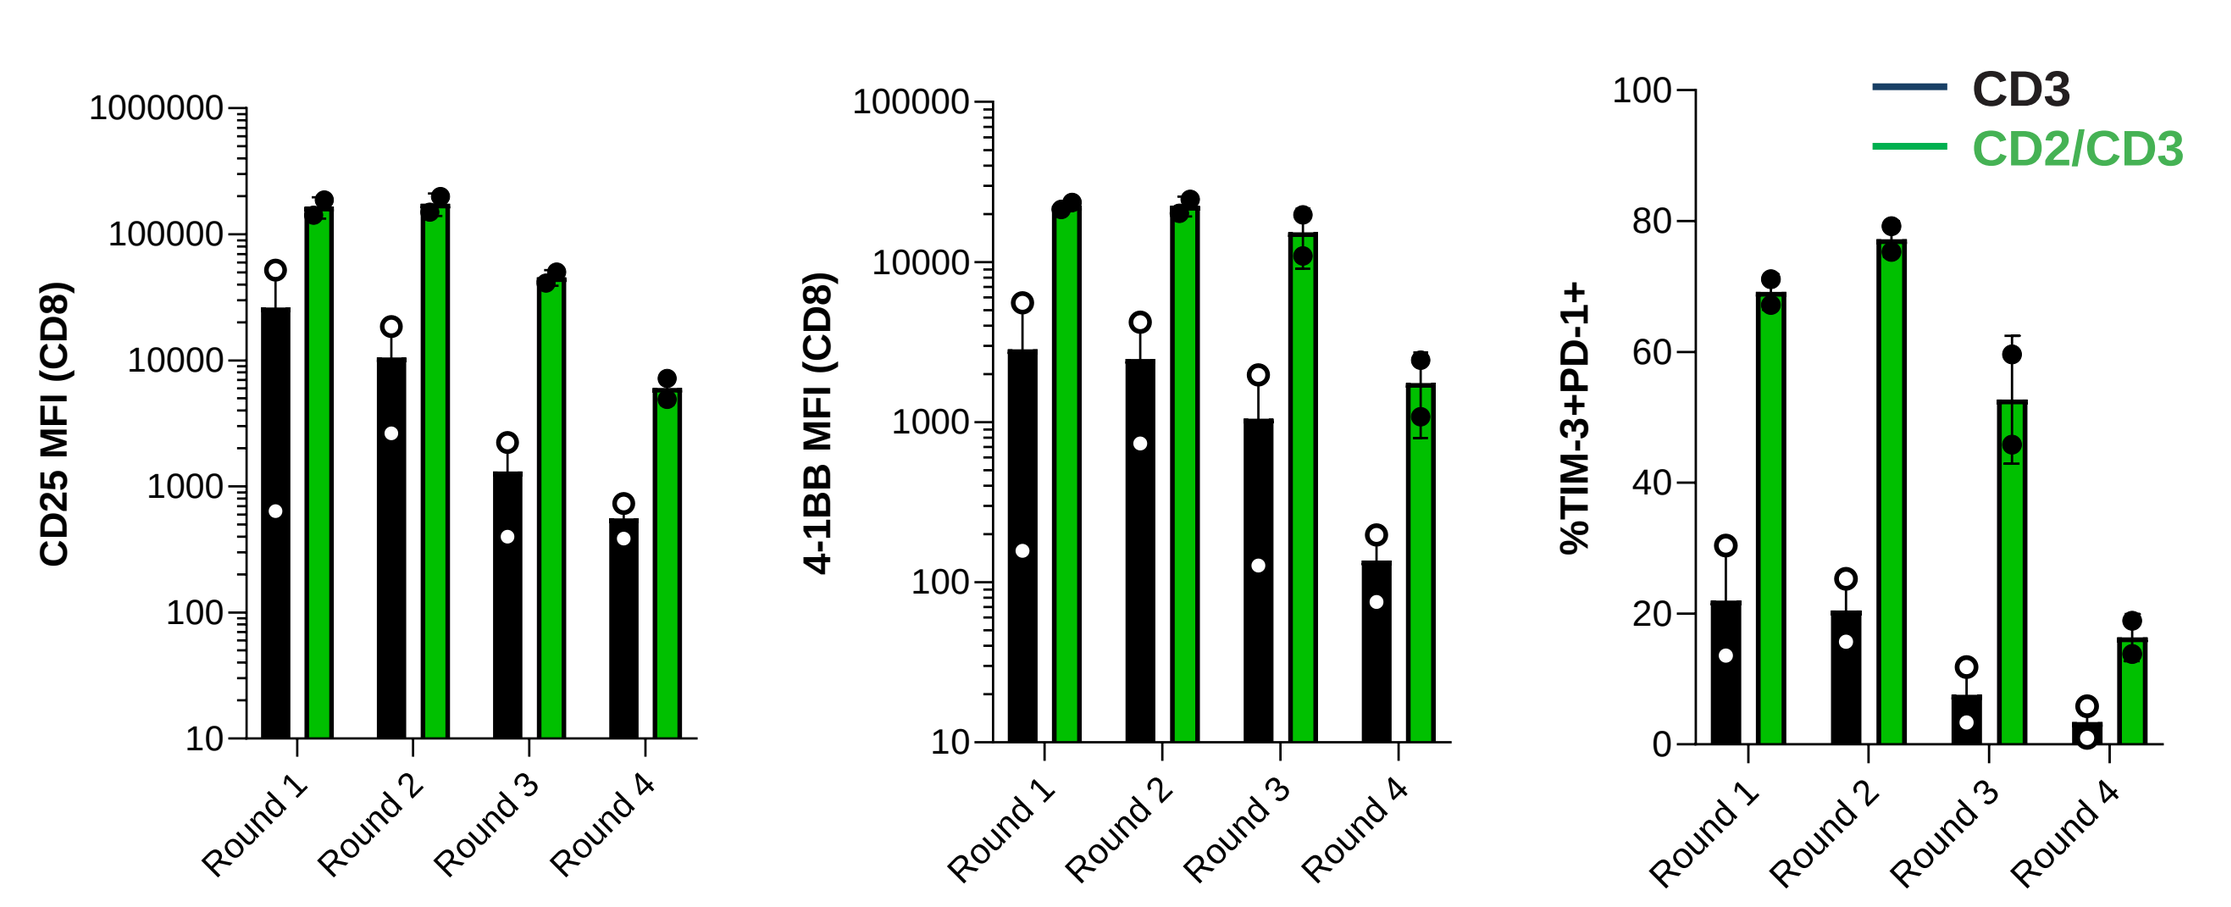
CD25 MFI (CD8)
4-1BB MFI (CD8)
%TIM-3+PD-1+
CD3
CD2/CD3
Round 1
Round 2
Round 3
Round 4

T cell cytotoxicity after 3 rounds of repetitive stimulation

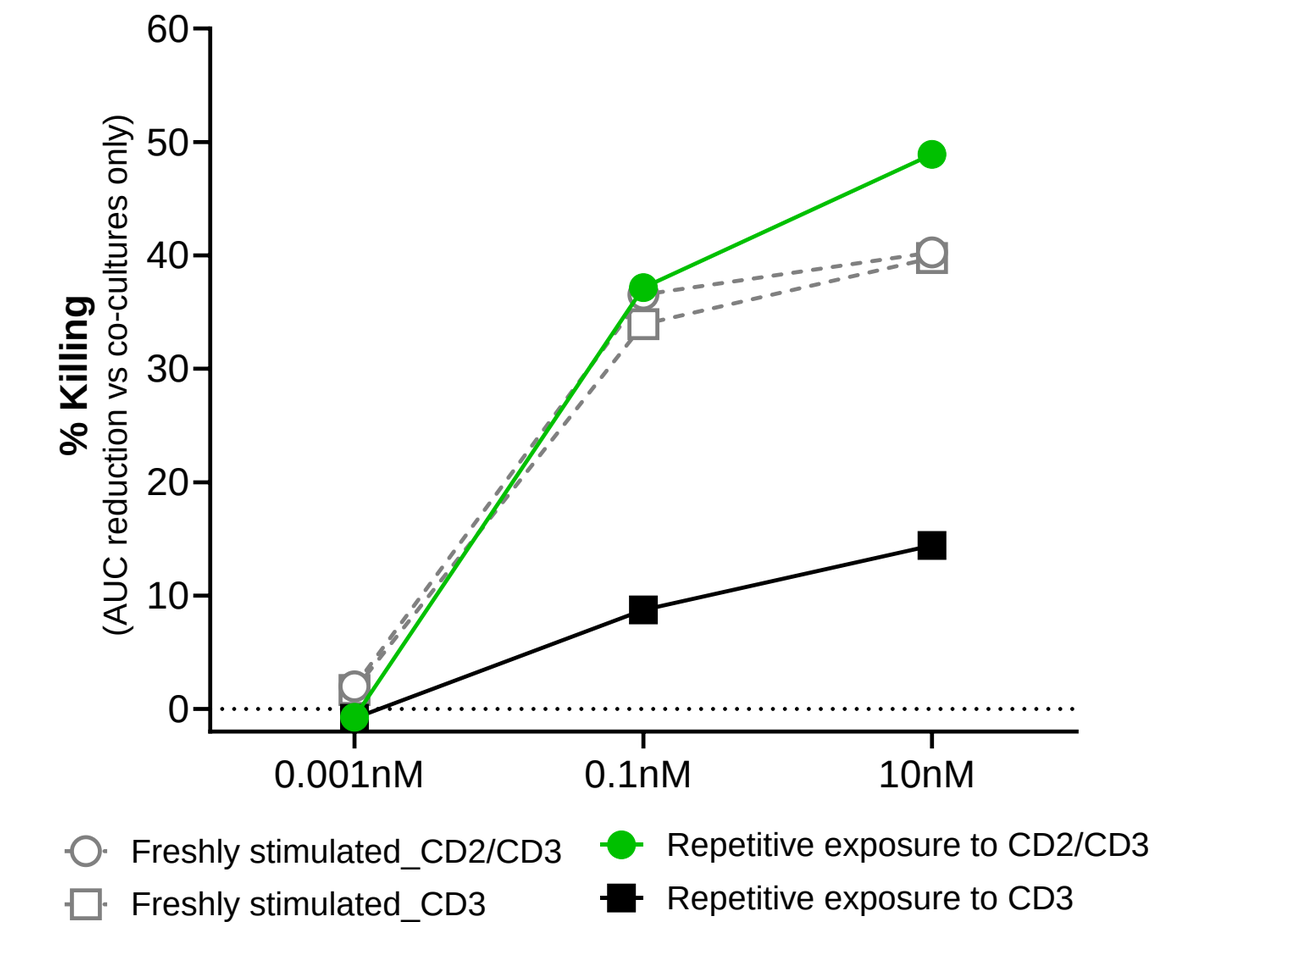
% Killing (AUC reduction vs co-cultures only)
0.001nM
0.1nM
10nM
Freshly stimulated_CD2/CD3
Freshly stimulated_CD3
Repetitive exposure to CD2/CD3
Repetitive exposure to CD3

T cells were repetitively exposed to DARPins captured by EpCAM and MSLN coated to the plates. After each round of stimulation, a portion of cells were taken for analysis by flow cytometry. After 3 rounds, T cells were co-cultured with NucLightRed (NLR) labeled Caov-3 cells and DARPins for 48h in an IncuCyte to measure the NLR signal over time.

Conclusions

We present a preclinical proof of concept for a multi-targeting DARPin T cell engager, with CD2 co-simulation and a TAA-based Switch to mask CD3 in the absence of TAAs. Our data show:

- ❖ Co-engagement of CD2 leads to sustained T cell activation and cytotoxic capacity, preventing dysfunction.
- ❖ Masking of CD3 allows higher specificity on the tumor, without compromising potency or needing to down-tune the CD3 affinity.
- ❖ CD2/CD3 Switch DARPin effectively induces potent tumor regression *in vivo,* whilst maintaining a good safety profile with reduced cytokine release compared to an unmasked CD3 with CD2 co-stimulation which ultimately leads to T cell depletion.

Acknowledgments – Special thanks to the Pre-clinical Platforms team: Amelie Croset, Marco Franchini, Alienor Auge, Norbert Fic, Yvonne Kaufmann, and Tamara Lekishvili
Disclosures: this research was funded by Molecular Partners AG, Zurich-Schlieren, Switzerland. All authors are employees and stock owners of Molecular Partners AG, Zurich-Schlieren, Switzerland

For any questions, please contact:
info@molecularpartners.com / attention of Marcela Guzman